816

DRP-104, A Novel Broad Acting Glutamine Antagonist, Induces Durable Anti-Tumor Responses In Vivo by Inhibiting Tumor Glutamine Addiction, Remodeling the Tumor Microenvironment and Stimulating Both the Innate & Adaptive Immune Systems

Yumi Yokoyama, Ph.D., Michael Nedelcovych, Ph.D. and Robert Wild, Ph.D.
Dracen Pharmaceuticals Inc., 780 Third Ave, 45th Floor, New York, NY 10017

Summary

Glutamine is a conditionally essential amino acid for rapidly proliferating cancer cells, thus depriving the same fuel from immune cells and contributing to tumor immune evasion. DRP-104 was designed as a novel prodrug of the broad acting glutamine antagonist 6-Diazo-5-oxo-L-norleucine (DON). DRP-104 is inert in its prodrug form, affords high levels of plasma and gastro-intestinal (GI) tissue stability; has high tumor cell permeability and preferential tumor versus plasma/GI tissue distribution for DON. Here we sought to (1) characterize *in vitro* biochemical and biological activities, (2) identify the immune-modulatory mechanism of action, and (3) evaluate *in vivo* anti-tumor efficacy of DRP-104 as a single agent. DRP-104 and its active moiety DON showed glutamine dependent inhibition of cancer cell growth *in vitro*, correlating with broad glutamine pathway inhibition. *In vivo*, DRP-104 is preferentially converted to DON in tumors and has minimal plasma/GI tissue distribution. Metabolomic profiling in tumor samples showed widespread changes indicative of disruption of tumor anabolism and canonical cancer metabolism pathways. In addition, glutamine and various other amino acids were significantly increased while several immune-suppressive metabolites were decreased. DRP-104 treatment resulted in substantial and broad changes in various immune cell infiltrates, such as increased TIL, T, NK and NKT cells. T cells were more proliferative and less exhausted. TAMs were polarized to M1 phenotype. DRP-104 treatment resulted in significant reduction in PD-L1 expression on tumors, macrophages. Pro-tumorigenic proteins, such as VEGF, KC (IL-8), and IL-10 were decreased. Finally, DRP-104 showed significant tumor growth inhibition including curative effects as a single agent in mouse syngeneic tumor models.

Background

Dracen's Glutamine Antagonist, DRP-104, Preferentially Delivers DON to Tumors Leading to Broad Glutamine Pathway Inhibition

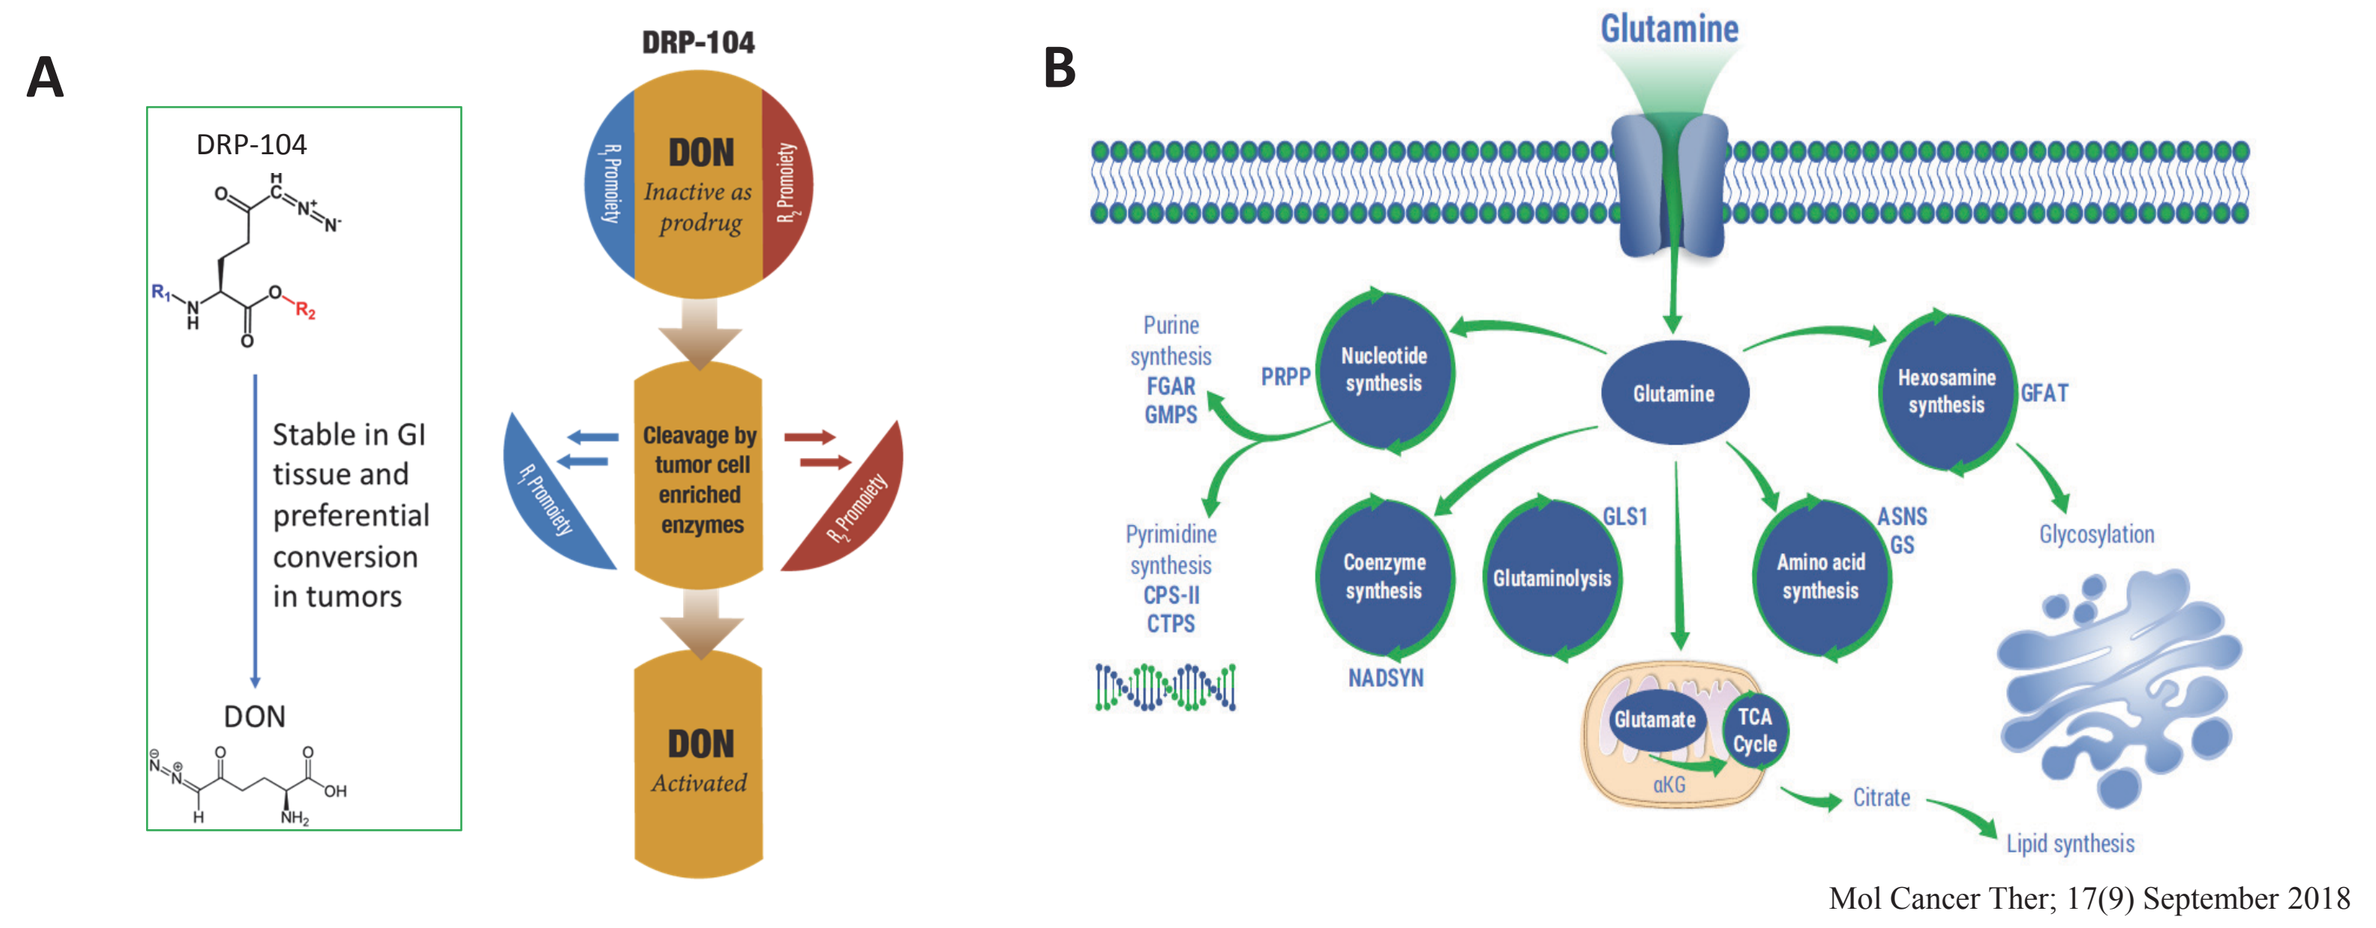
A
B
DRP-104
Stable in GI tissue and preferential conversion in tumors
DON
DON Inactive as prodrug
Cleavage by tumor cell enriched enzymes
DON Activated
Glutamine
Purine synthesis FGAR GMPS
PRPP
Nucleotide synthesis
Hexosamine synthesis
GFAT
Pyrimidine synthesis CPS-II CTPS
Coenzyme synthesis
Glutaminolysis
GLS1
Amino acid synthesis
ASNS GS
Glycosylation
NADSYN
Glutamate
TCA Cycle
αKG
Citrate
Lipid synthesis
Mol Cancer Ther; 17(9) September 2018

Figure 1. A. DRP-104 is a prodrug of the broad acting glutamine antagonist DON (6-Diazo-5-oxo-L-norleucine). DRP-104 is inactive in its prodrug form with high plasma and GI tissue stability. DRP-104 is preferentially distributed in tumors where it is biotransformed and activated to the active moiety DON. **B.** Multiple glutamine dependent pathways are inhibited by DON. Enzymes targeted by DON are shown in blue bold letters.

DRP-104 and DON Demonstrate Glutamine Dependent Effects on Cell Viability *In Vitro*

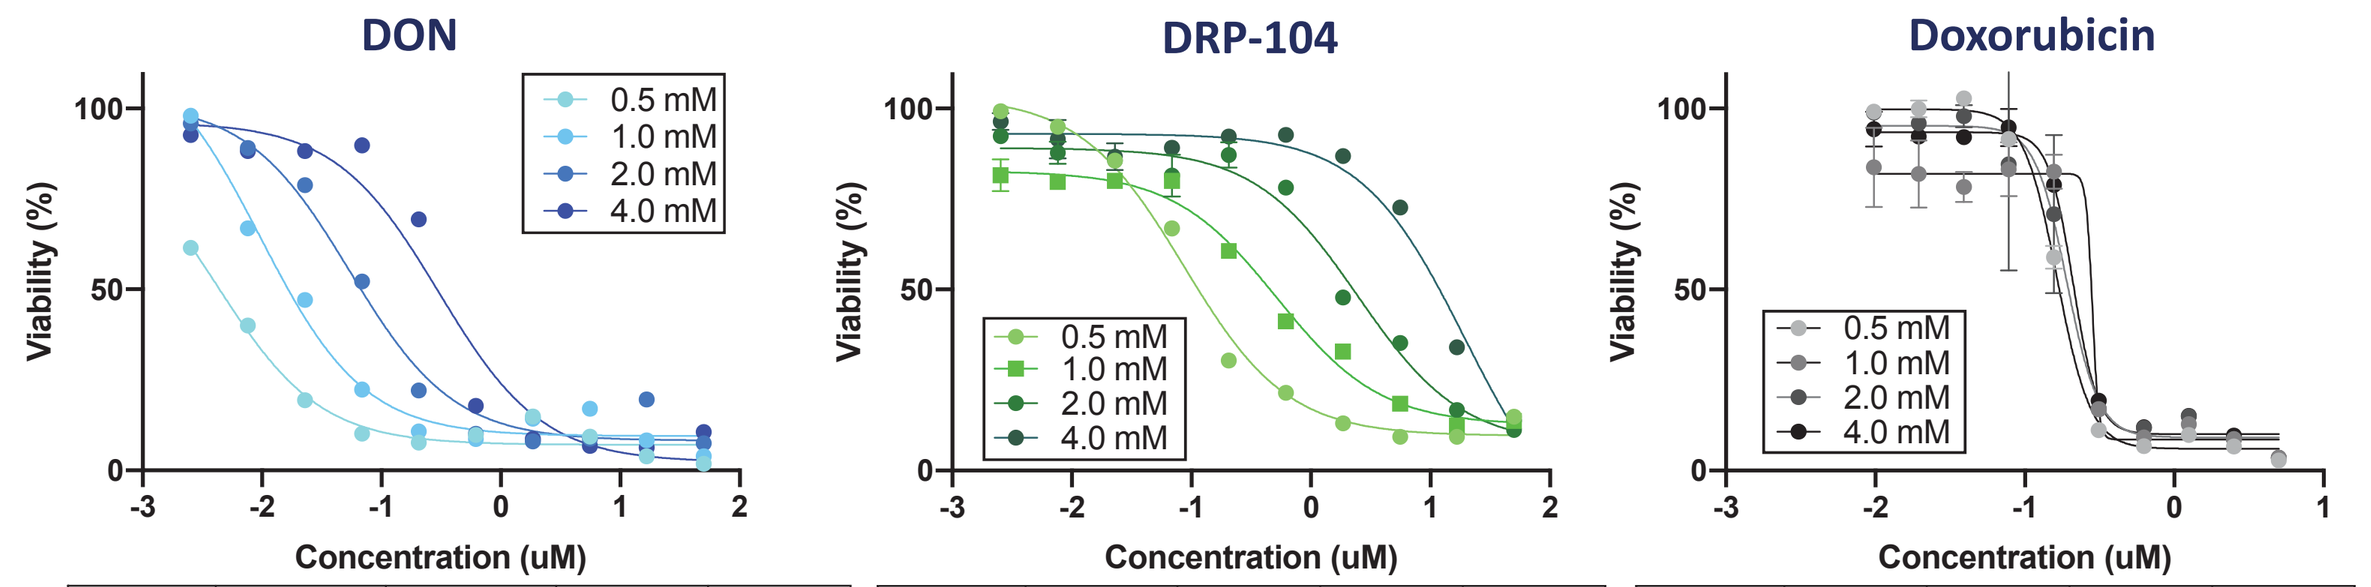
DON
DRP-104
Doxorubicin
0.5 mM
1.0 mM
2.0 mM
4.0 mM
Viability (%)
Concentration (uM)

Gln	0.5 mM	1.0 mM	2.0 mM	4.0 mM
IC50	0.004200	0.009670	0.05439	0.3016

Gln	0.5 mM	1.0 mM	2.0 mM	4.0 mM
IC50	0.08627	0.5198	2.479	19.32

Gln	0.5 mM	1.0 mM	2.0 mM	4.0 mM
IC50	0.1732	0.2070	0.1988	0.2198

Figure 2. MC38 mouse colon carcinoma cells were plated in media with different concentrations of glutamine (0.5, 1.0, 2.0, and 4.0 mM), and were treated with DON, DRP-104 or Doxorubicin for 3 days. Cell viability was measured by Cell Titer Glo (Promega).
Both DON and DRP-104, but not doxorubicin showed glutamine concentration dependent inhibition on cell viability.

DRP-104 Achieves 6x Tumor/Plasma and 12x Tumor/Jejunum Exposure Ratios for DON in CES-1 KO Mice Providing PoC for Prodrug Tumor Targeting *In Vivo*

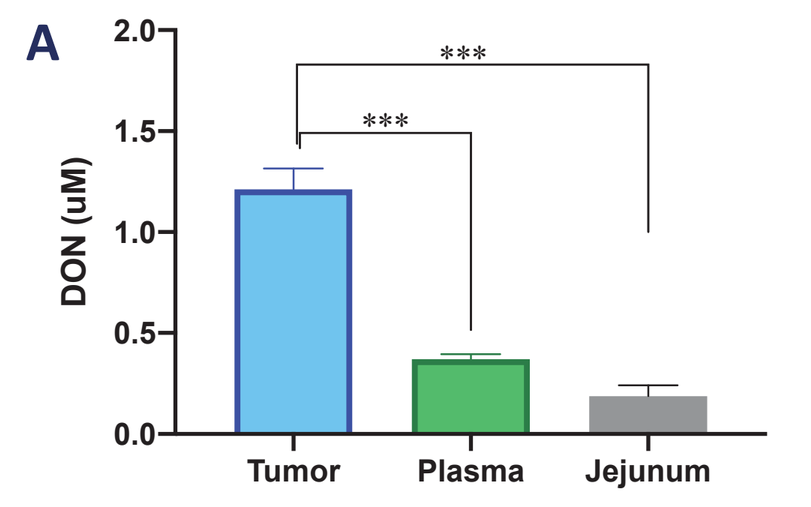
A
DON (uM)

Tumor
Plasma
Jejunum

B

$AUC_{0-\infty}$ Ratio

Tumor/Plasma	Tumor/Jejunum
6.6	12

Figure 3. EL4 mouse lymphoma bearing CES-1 ko mice (C57BL/6 CES1-/-) were dosed *s.c.* with DRP-104 (2.6 mg/kg). Tumor, plasma and jejunum were harvested 0.25-6 hr after the administration to calculate $AUC_{0-\infty}$. $AUC_{0-\infty}$ (nmol.h/ml) was calculated by the macro PK Functions for Microsoft Excel. **A.** DON concentration in tumor, plasma or Jejunum 30 min after dosing. **B.** $AUC_{0-\infty}$ ratio. ***: p<0.001.

DRP-104 Administration Yields Dose-Dependent Changes in Key Pharmacodynamic Metabolites

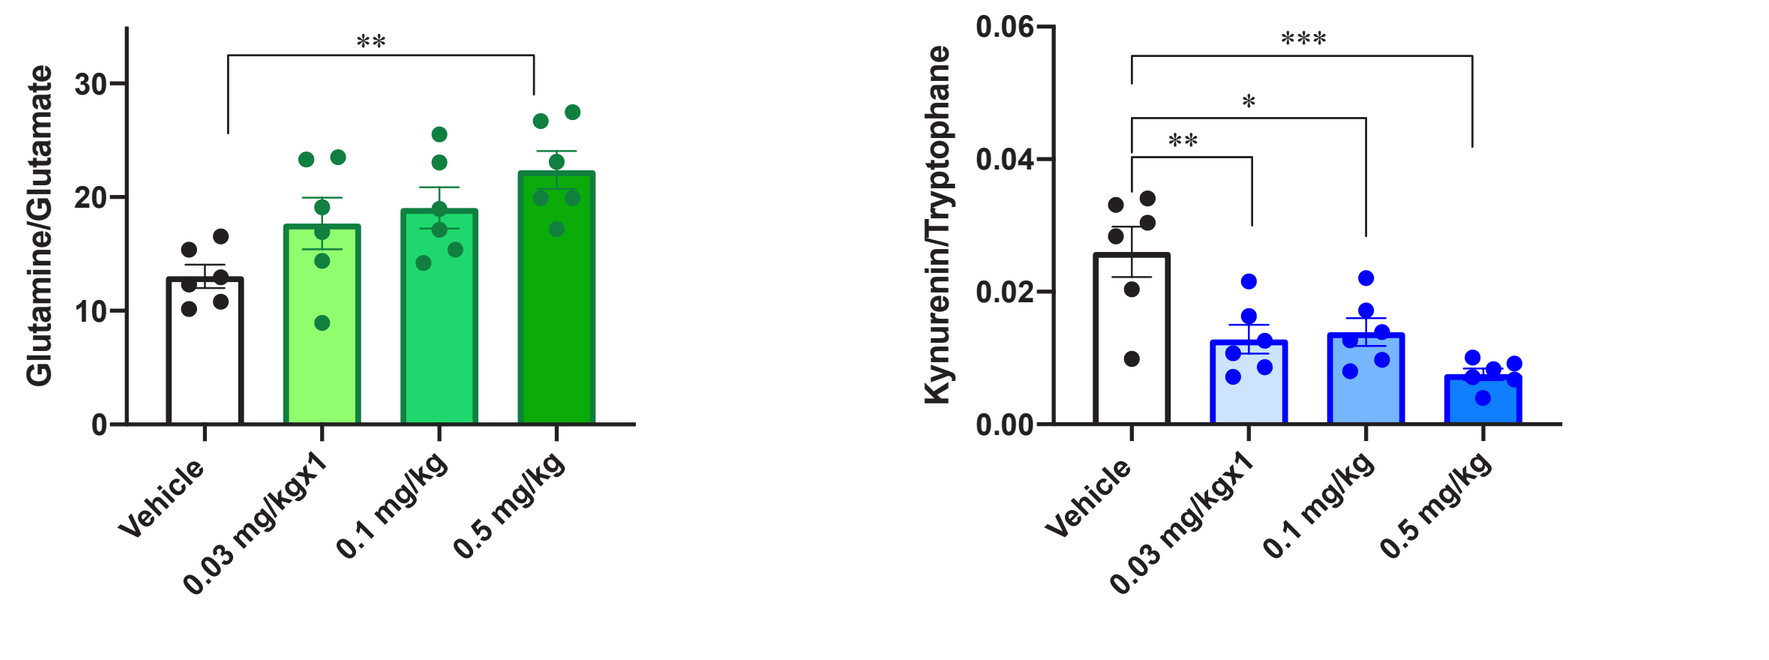
Glutamine/Glutamate
Kynurenine/Tryptophan
Glutamine/Glutamate
Kynurenin/Tryptophane
Vehicle
0.03 mg/kgx1
0.1 mg/kg
0.5 mg/kg

Figure 4. MC38 tumor bearing animals were dosed s.c. with DRP-104 for 5 consecutive days.
Consistent and significant dose-dependent changes in plasma pharmacodynamic metabolites demonstrating target engagement of DRP-104. Modulation of Kynurenine/Tryptophan metabolism suggests DRP-104 may have additional impact on immune cell function in the TME. **: p<0.01; ***: p<0.001.

DRP-104 Treatment Results in Broad Metabolomic Reprogramming of the TME Providing Favorable Conditions for Immune Cell Function

Summary Counts Welch Two Sample t-Test	Low QD / Veh QD	High QD / Veh QD
Total number of biochemicals detected		
Total number of biochemicals with p≤0.05	157	359
Biochemicals (↑\|↓)	91 \| 66	188 \| 171
%Total biochemicals significantly different	18%	41%

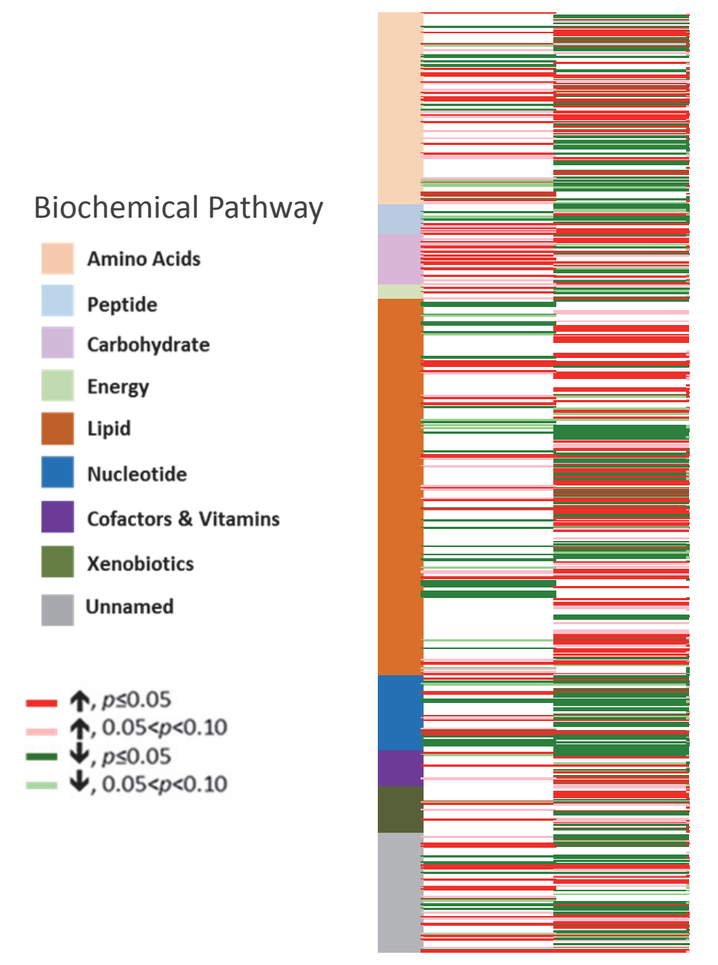
Biochemical Pathway
Amino Acids
Peptide
Carbohydrate
Energy
Lipid
Nucleotide
Cofactors & Vitamins
Xenobiotics
Unnamed
↑, p≤0.05
↑, 0.05<p<0.10
↓, p≤0.05
↓, 0.05<p<0.10

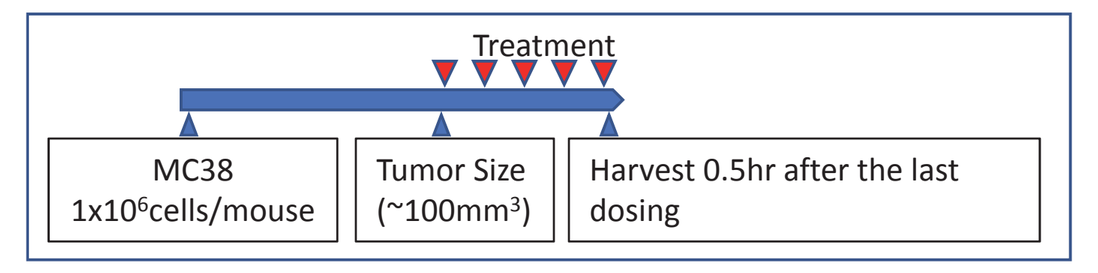
Treatment
MC38
1x10⁶cells/mouse
Tumor Size
(~100mm³)
Harvest 0.5hr after the last dosing

DRP-104 treatment (0.03 or 0.5 mg/kg, *sc, q.d.* x5) resulted in significant dose dependent changes in various metabolites in the TME compared to vehicle:

- Inhibition of glutaminolysis leading to increased glutamine
- Decreased nucleotide metabolites
- Increased amino acid metabolites
- Decreased kynurenine levels
- Altered glutathione metabolism
- Decreased levels of COX/LOX metabolites such as PGE2
- Increased glucose and fructose levels
- Decreased Glycosylation precursors

Figure 5. MC38 tumor bearing mice were treated with DRP-104 (Low QD: 0.03 or High QD: 0.5 mg/kg) or vehicle *s.c., q.d.* for 5 days. Tumors were obtained 30 min after the last dose and the lysates were analyzed by LC/MS. 869 biochemical metabolites were analyzed and quantified. Graph shows summary of statistical differences and pathway heat map overview. For the heat map red ($p \le 0.05$) and light red ($0.05 < p < 0.10$) represent significantly increased and trending increased biochemicals, respectively. Green ($p \le 0.05$) and light green ($0.05 < p < 0.10$) represent significantly decreased and trending decreased biochemicals, respectively.

DRP-104 Treatment *In Vivo* Results in Dramatic Modulation of Multiple Relevant Tumor and Immune Cell Scores as Measured by Nanostring IO360

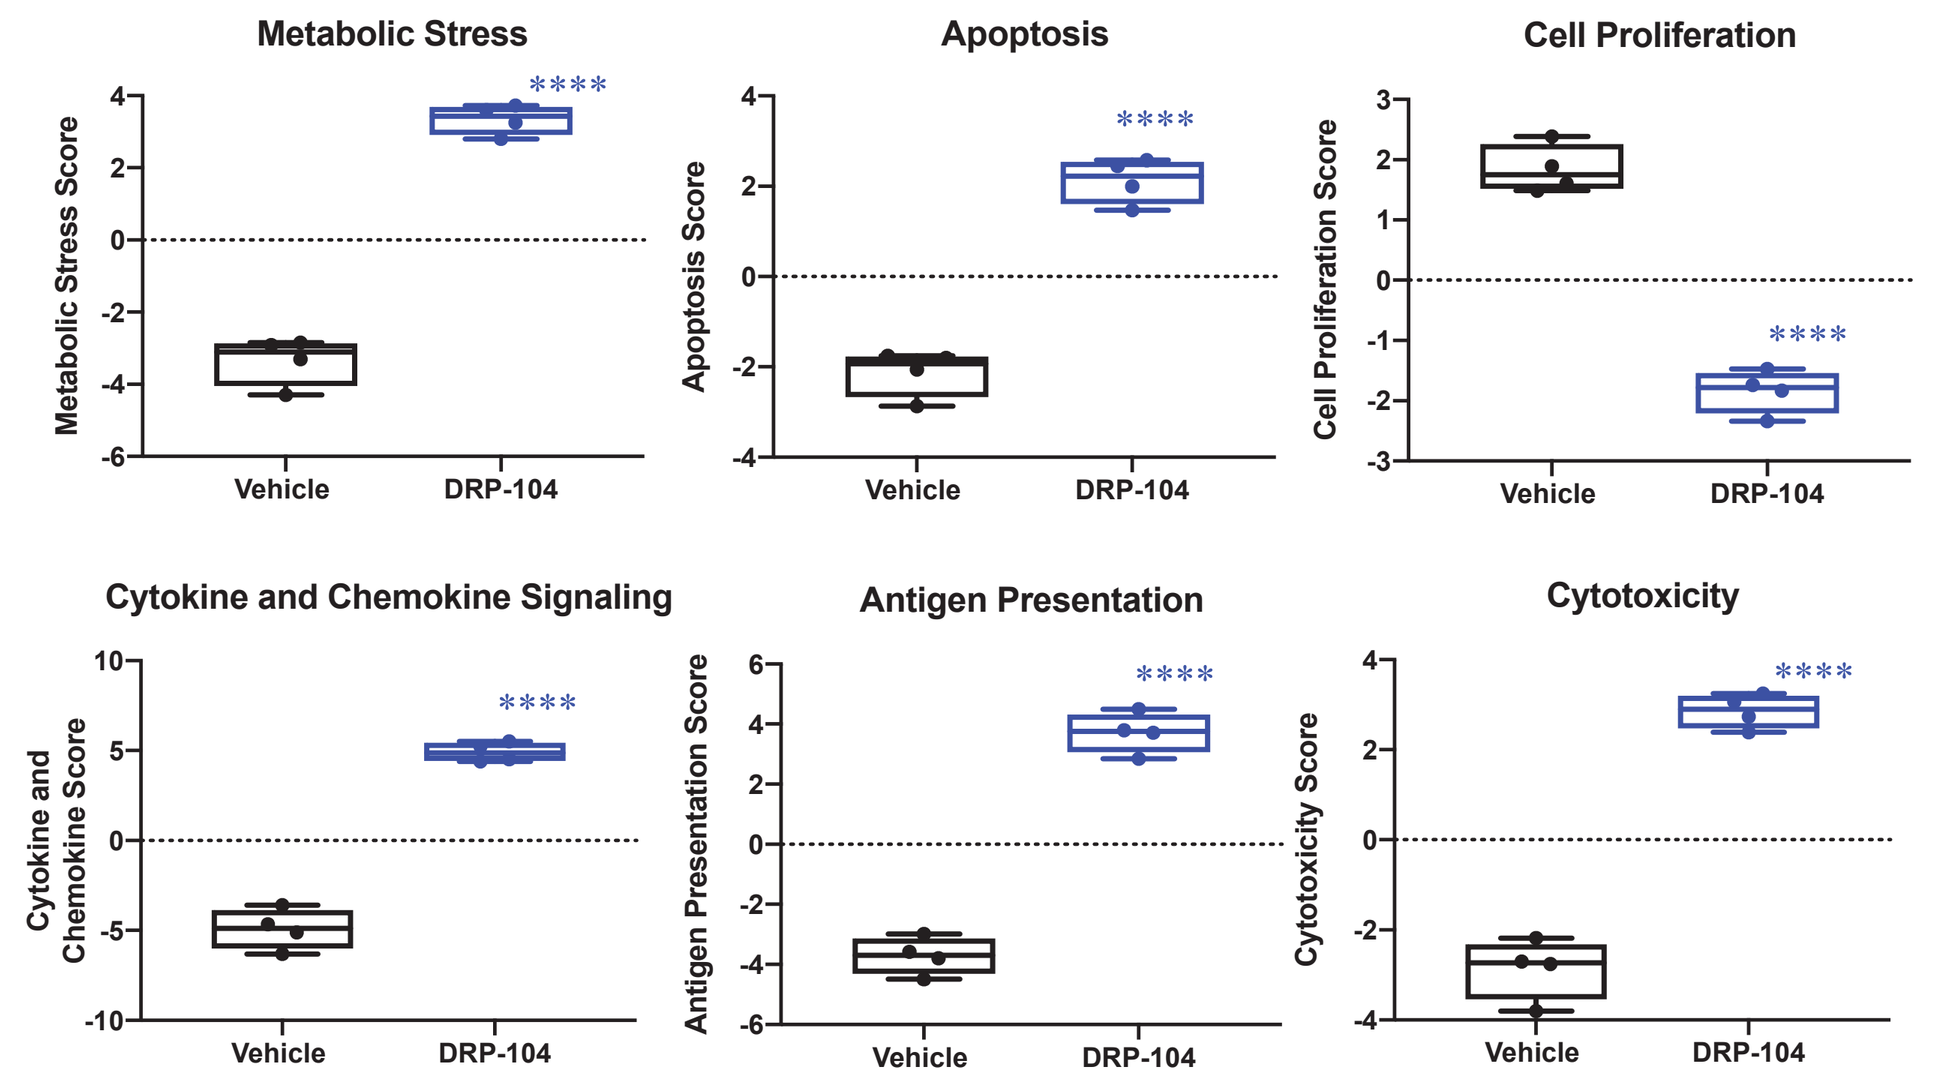
Metabolic Stress
Apoptosis
Cell Proliferation
Cytokine and Chemokine Signaling
Antigen Presentation
Cytotoxicity
Vehicle
DRP-104

Figure 6. MC38 cells were inoculated *s.c.* in C57BL/6 mice. When tumor size reached ~400 mm³, mice were treated with DRP-104 (0.5 mg/kg) or vehicle *s.c., q.d.* for 5 days. mRNA was extracted from tumors and analyzed by Nanostring IO360 Panel. ****: p<0.0001.
DRP-104 treatment was associated with dramatic modulation of multiple relevant tumor and immune cell scores suggesting direct anti-tumor effects while stimulating immune cell function.

DRP-104 Treatment Increases Activation and Infiltration of Lymphocytes into Tumors *In Vivo*

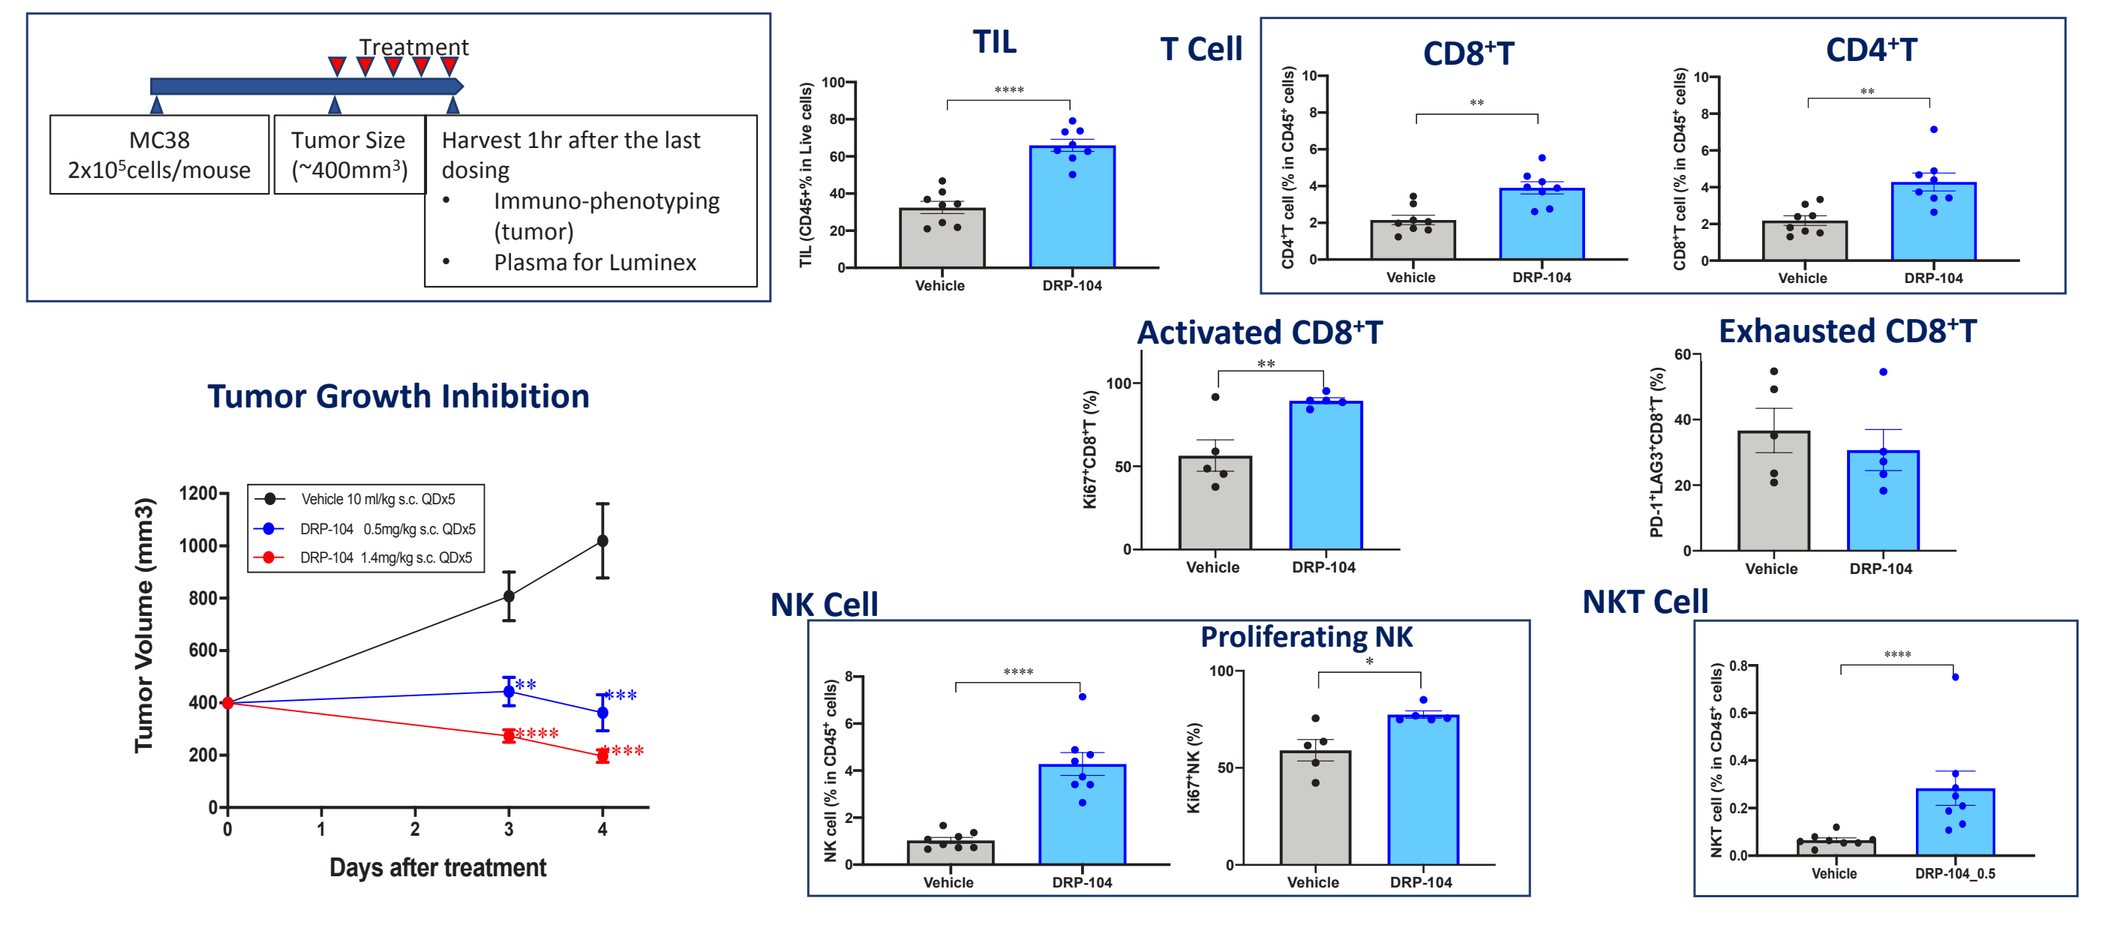
Treatment
MC38
2x10⁵cells/mouse
Tumor Size
(~400mm³)
Harvest 1hr after the last dosing
Immuno-phenotyping (tumor)
Plasma for Luminex
TIL
T Cell
CD8⁺T
CD4⁺T
Activated CD8⁺T
Exhausted CD8⁺T
Tumor Growth Inhibition
Tumor Volume (mm3)
Days after treatment
NK Cell
Proliferating NK
NKT Cell
Vehicle
DRP-104

Figure 7. MC38 cells were inoculated *s.c.* in C57BL/6 mice. When tumor size reached ~400 mm³, mice were treated with DRP-104 (0.5 or 1.4 mg/kg) or vehicle *s.c., q.d.* for 5 days. Phenotypic analyses of tumor infiltrating immune cells and tumor cells with DRP-104 (0.5 mg/kg) were performed by flow cytometry. *: p<0.05; **: p<0.01; ***: p<0.001; ****: p<0.0001.
Increased tumor infiltration of activated T, NK and NKT cells was associated with anti-tumor activity suggesting DRP-104's MOA includes immuno-oncology effects.

DRP-104 Treatment Increases Polarized Macrophage Infiltration into Tumors *In Vivo*

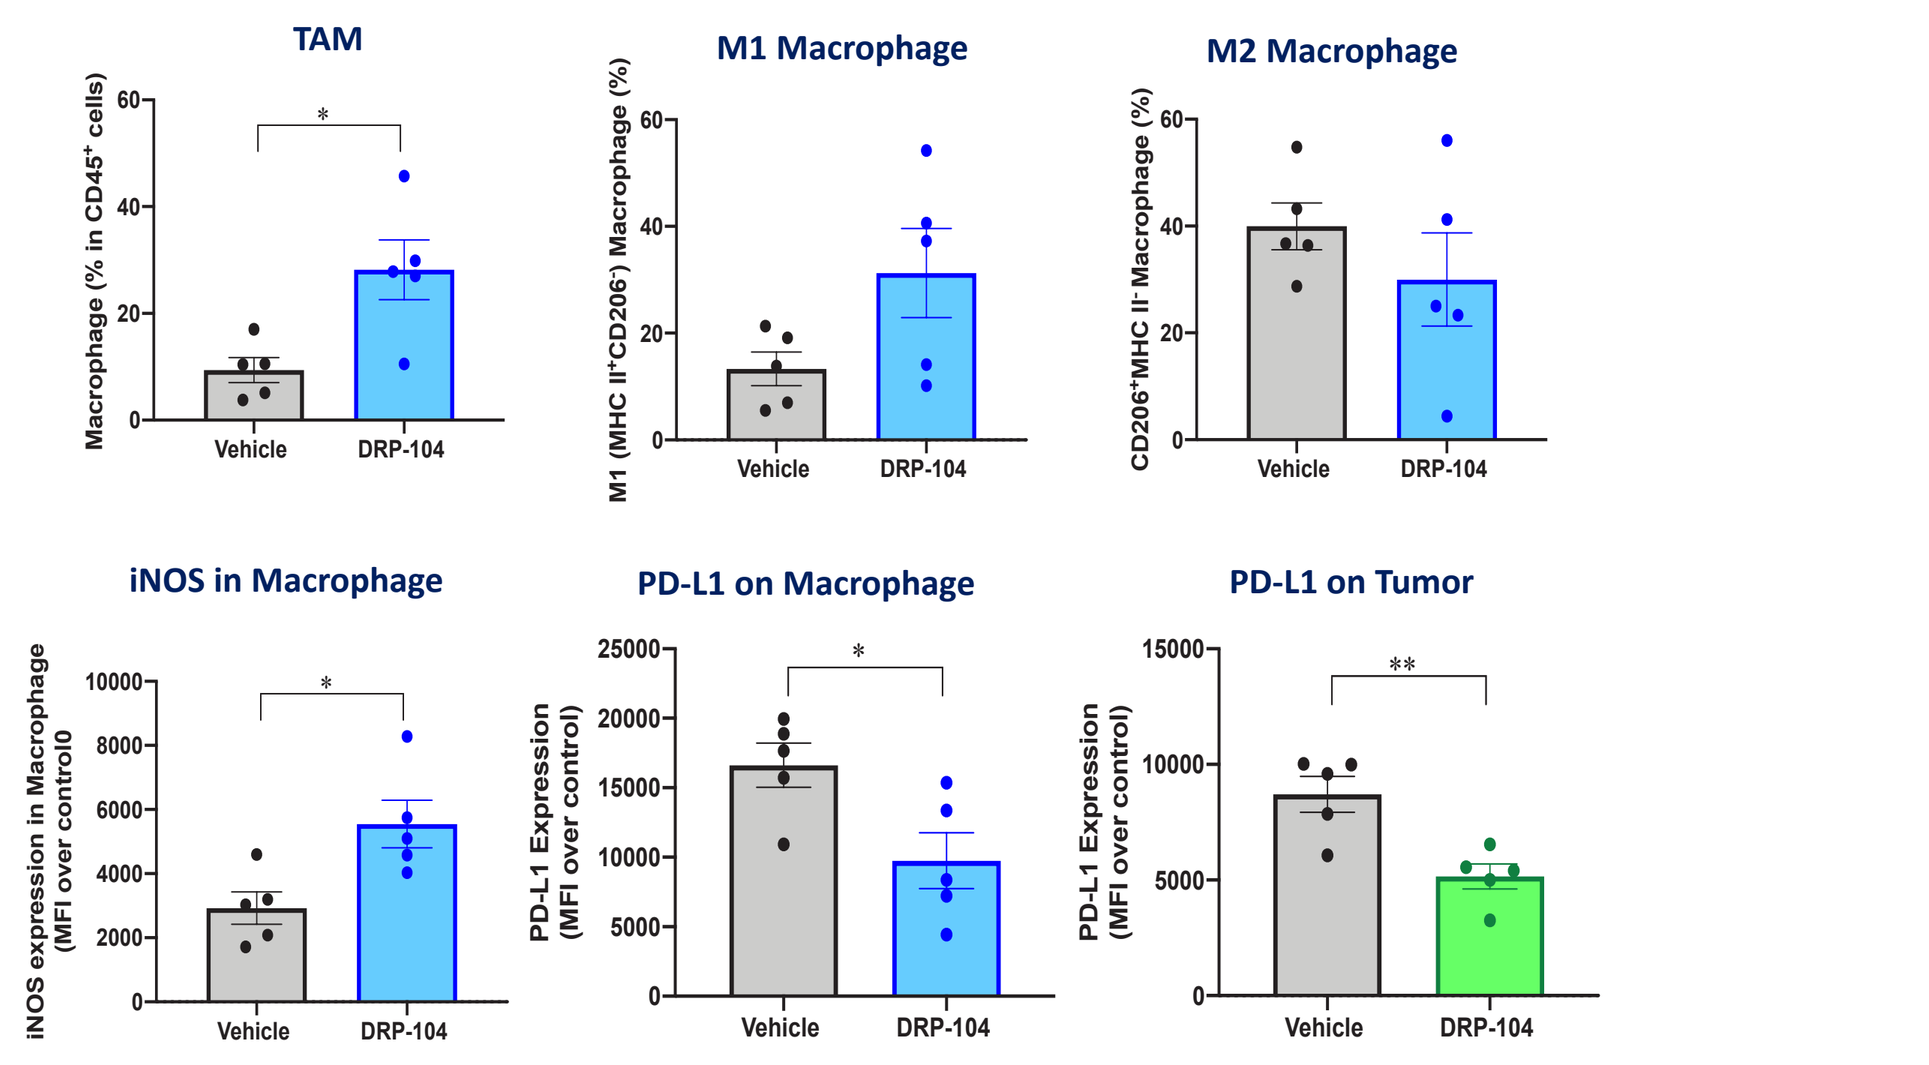
TAM
M1 Macrophage
M2 Macrophage
iNOS in Macrophage
PD-L1 on Macrophage
PD-L1 on Tumor
Vehicle
DRP-104

Figure 8. MC38 cells were inoculated *s.c.* in C57BL/6 mice. When tumor size reached ~400 mm³, mice were treated with DRP-104 (0.5mg/kg) or vehicle *s.c.*, *q.d.* for 5 days. Phenotypic analyses of tumor infiltrating immune cells and tumor cells were performed by flow cytometry. *:p<0.05.
Increased tumor infiltration of M1 polarized macrophages and decreased expression of PD-L1 were associated with anti-tumor activity.

DRP-104 Modulates *In Vivo* Cytokines in TME toward Anti-tumor Phenotype

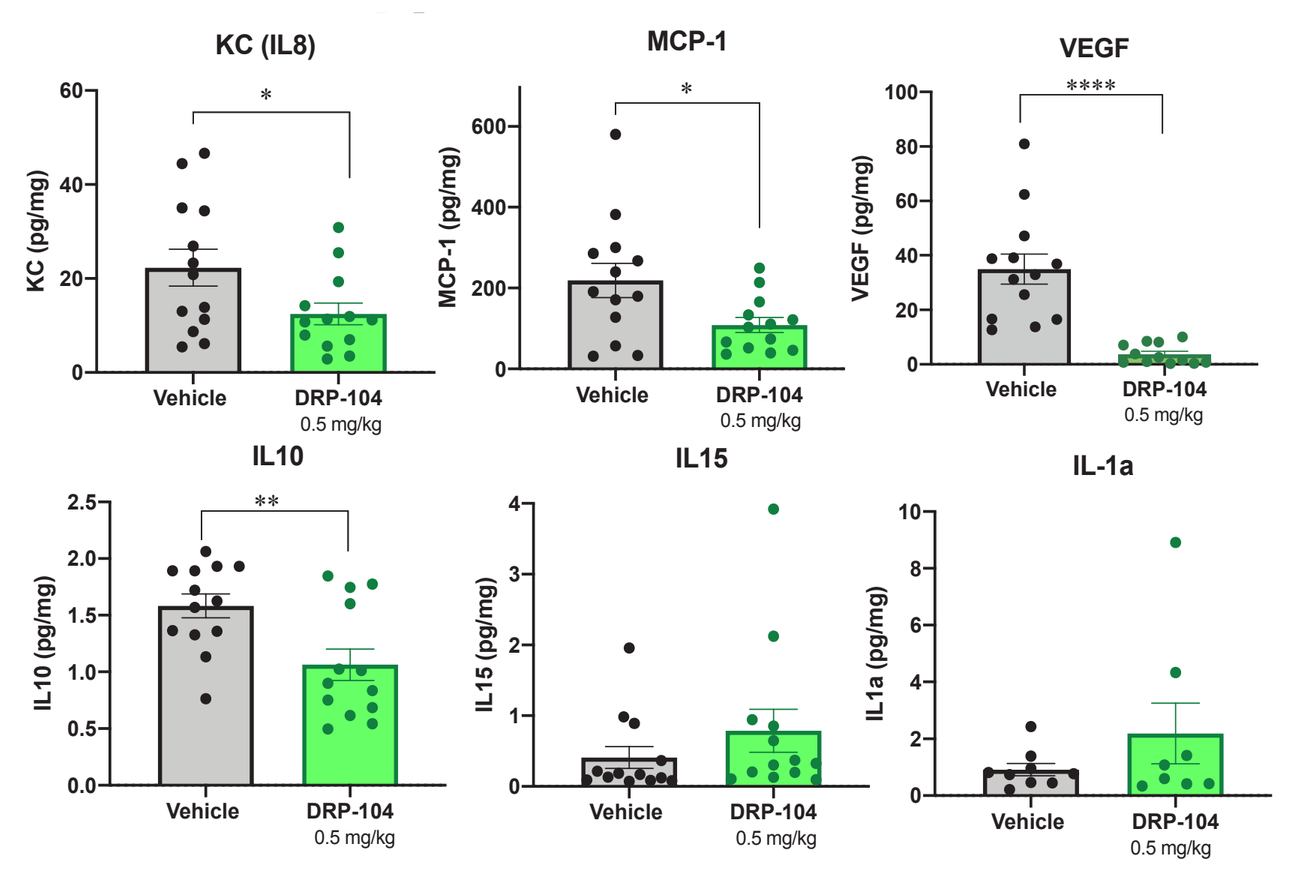
KC (IL8)
MCP-1
VEGF
IL10
IL15
IL-1a
Vehicle
DRP-104
0.5 mg/kg

Figure 9. Cytokines in tumor lysates from MC38 study shown in Figure 6 and 7 were measured by Luminex.
Data indicate mean+/- SEM. *: p<0.05; **: p<0.01; ****: p<0.0001.

DRP-104 Showed Dose Dependent Anti-Tumor Efficacy and Survival Including Long-Term Durable Responses in MC38 Colon Cancer Model

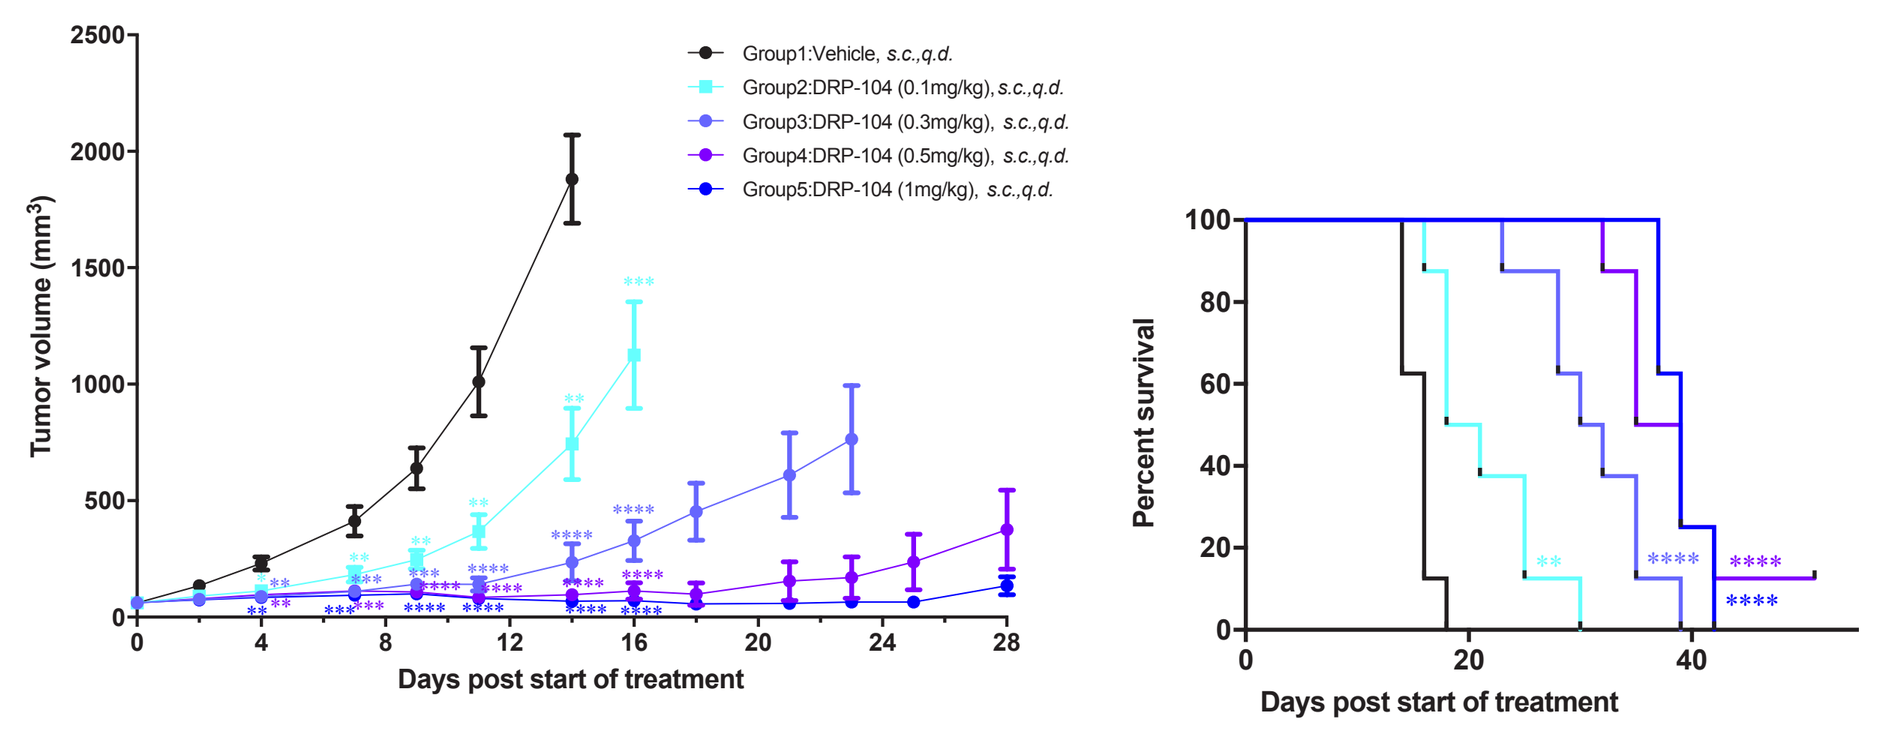
Group1:Vehicle, s.c.,q.d.
Group2:DRP-104 (0.1mg/kg), s.c.,q.d.
Group3:DRP-104 (0.3mg/kg), s.c.,q.d.
Group4:DRP-104 (0.5mg/kg), s.c.,q.d.
Group5:DRP-104 (1mg/kg), s.c.,q.d.
Tumor volume (mm³)
Days post start of treatment
Percent survival
Days post start of treatment

Figure 10. MC38 cells were inoculated *s.c.* in C57BL/6 mice. When tumor size reached ~100 mm³, mice were treated with DRP-104 or vehicle *s.c., q.d.* for 28 days. *: p<0.05; **: p<0.01; ***: p<0.001; ****: p<0.0001.

Conclusions and Mechanism of Action of DRP-104

- ❖ Both DRP-104 and the active form DON showed Glutamine dependent inhibition of *in vitro* cancer cell growth.
- ❖ DRP-104 achieved 6x tumor/plasma and 12x tumor/jejunum ratios for DON in CES-1 ko mice proving successful partitioning of prodrug to direct DON, the active moiety, to tumors.
- ❖ DRP-104 treatment yielded dose-dependent changes in key plasma metabolites, broad metabolomic reprogramming of the TME, and dramatic immuno-oncology related gene modulations.
- ❖ Immuno-phenotypical analysis showed that DRP-104 induced substantial and broad changes in various immune cell infiltrates, such as increased TIL, T, NK and NKT cells. T cells were more proliferative and less exhausted, and TAMs were polarized to M1 phenotype. DRP-104 treatment resulted in significant reduction in PD-L1 expression on tumors, macrophages. All of these changes suggest broad remodeling of the TME to enhance the direct anti-tumor efficacy of DRP-104.

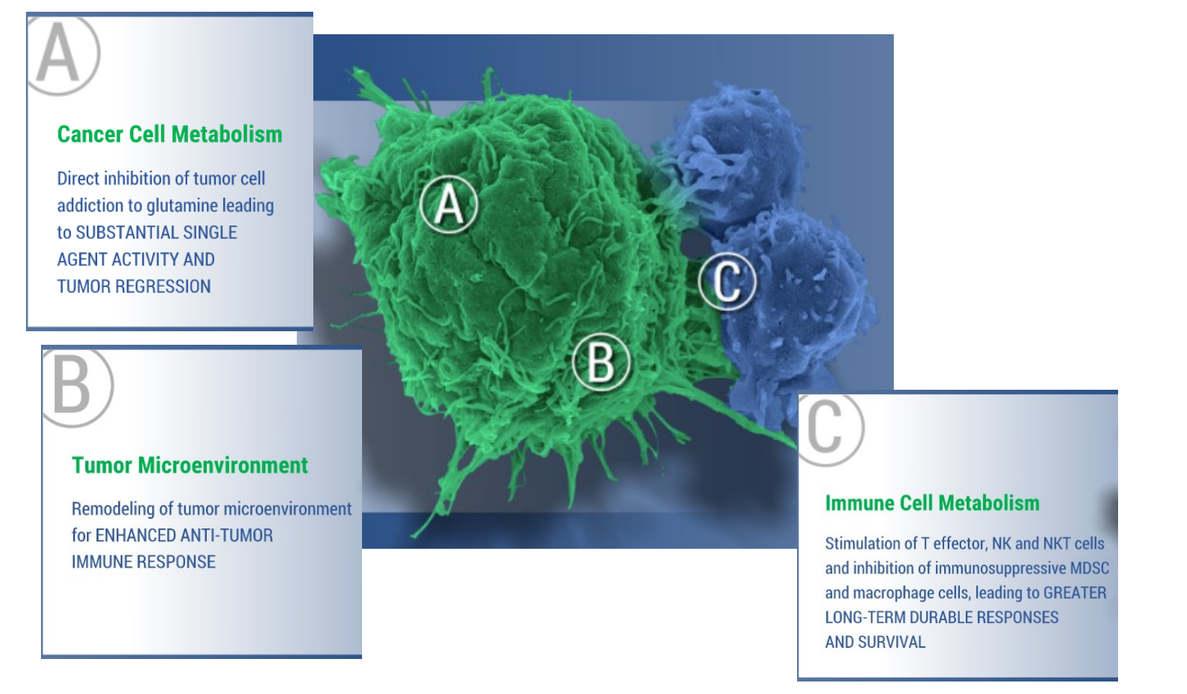
A
Cancer Cell Metabolism
Direct inhibition of tumor cell addiction to glutamine leading to SUBSTANTIAL SINGLE AGENT ACTIVITY AND TUMOR REGRESSION
B
Tumor Microenvironment
Remodeling of tumor microenvironment for ENHANCED ANTI-TUMOR IMMUNE RESPONSE
C
Immune Cell Metabolism
Stimulation of T effector, NK and NKT cells and inhibition of immunosuppressive MDSC and macrophage cells, leading to GREATER LONG-TERM DURABLE RESPONSES AND SURVIVAL

- ❖ Pro-tumorigenic proteins, such as VEGF, KC (IL-8), and IL-10 were decreased by DRP-104.
- ❖ Finally, DRP-104 inhibited tumor growth in a dose dependent manner, leading to significantly extended survival including long term durable cures.
- ❖ This data support the clinical development of DRP-104 as a single agent based on direct effects on tumor metabolism as well as broad modulation of anti-tumor immune response mechanisms.

Acknowledgement

We acknowledge Dr. Slusher and Dr. Rais (Johns Hopkins University) for PK analysis of DRP-104 in CES ko mice.